Fungal Eradication With Extended Rezafungin Treatment In a Mouse Model Of *Pneumocystis* Pneumonia

Taylor Sandison[1], Alan Ashbaugh[2], Melanie T. Cushion[2,3]

[1]Cidara Therapeutics, Inc, San Diego, CA, USA; [2]University of Cincinnati College of Medicine, Cincinnati, OH, USA;
[3]Cincinnati VAMC, Cincinnati, OH, USA

Taylor Sandison, MD, MPH
Cidara Therapeutics, Inc.
6310 Nancy Ridge Drive, Suite 101
San Diego, CA, USA
tsandison@cidara.com

21st ICHS Symposium on Infections in the Immunocompromised Host

INTRODUCTION

- *Pneumocystis* pneumonia has substantial impact on morbidity and mortality among immunocompromised populations, such as patients with haematological malignancies and stem cell transplant recipients[1]
- Trimethoprim/sulfamethoxazole is the current recommended agent for treatment and prevention of *Pneumocystis*, but safety and tolerability can limit its use and alternatives are lacking[1,2]
- Rezafungin is a novel, next-generation echinocandin that has demonstrated efficacy against *Pneumocystis* in vivo[4,5] and is being studied in an ongoing Phase 3 trial (ReSPECT; NCT04368559) for prevention of invasive fungal disease (IFD) caused by *Candida*, *Aspergillus*, and *Pneumocystis* in patients undergoing blood or marrow transplantation
- As previous studies show that current echinocandins deplete asci (sexual reproduction) of *Pneumocystis* but that nuclei (asexual reproduction) persist,[3] this study was conducted to evaluate whether nuclei are sustained or eliminated during rezafungin treatment

METHODS

The effects of rezafungin on *Pneumocystis* administered for an extended period, up to 8 weeks, was evaluated in immunosuppressed mice. C3H/HeN mice were infected with *P. murina* by exposure to seed mice with fulminant infection. Mice were administered vehicle as a negative control (C/S) or rezafungin 20 mg/kg 3x/week intraperitoneally starting at Week 6 post-infection. Drug administration was stopped after 2, 4, 6, and 8 weeks of treatment (at Weeks 8, 10, 12, and 14). Mice were then euthanized, and lungs were processed. Nuclei and asci counts for each lung were log transformed and statistical significance (p <0.05) was determined by analysis of variance (ANOVA); individual groups were compared by the Student- Newman-Keuls t test for multiple comparisons.

RESULTS

- After 2 weeks of treatment (at Week 8), rezafungin significantly reduced both nuclei and asci burdens compared with C/S (Figure)
- No asci were observed in rezafungin-treated mice at any timepoint
- Nuclei counts in rezafungin-treated mice were significantly reduced at all timepoints compared with Week 8–controls and at Weeks 10 and 12 compared with the previous week timepoint (Week 8 and Week 10, respectively). No nuclei were observed at Week 14.

Figure. Log_{10} mean nuclei and asci counts after 2-8 weeks of rezafungin treatment

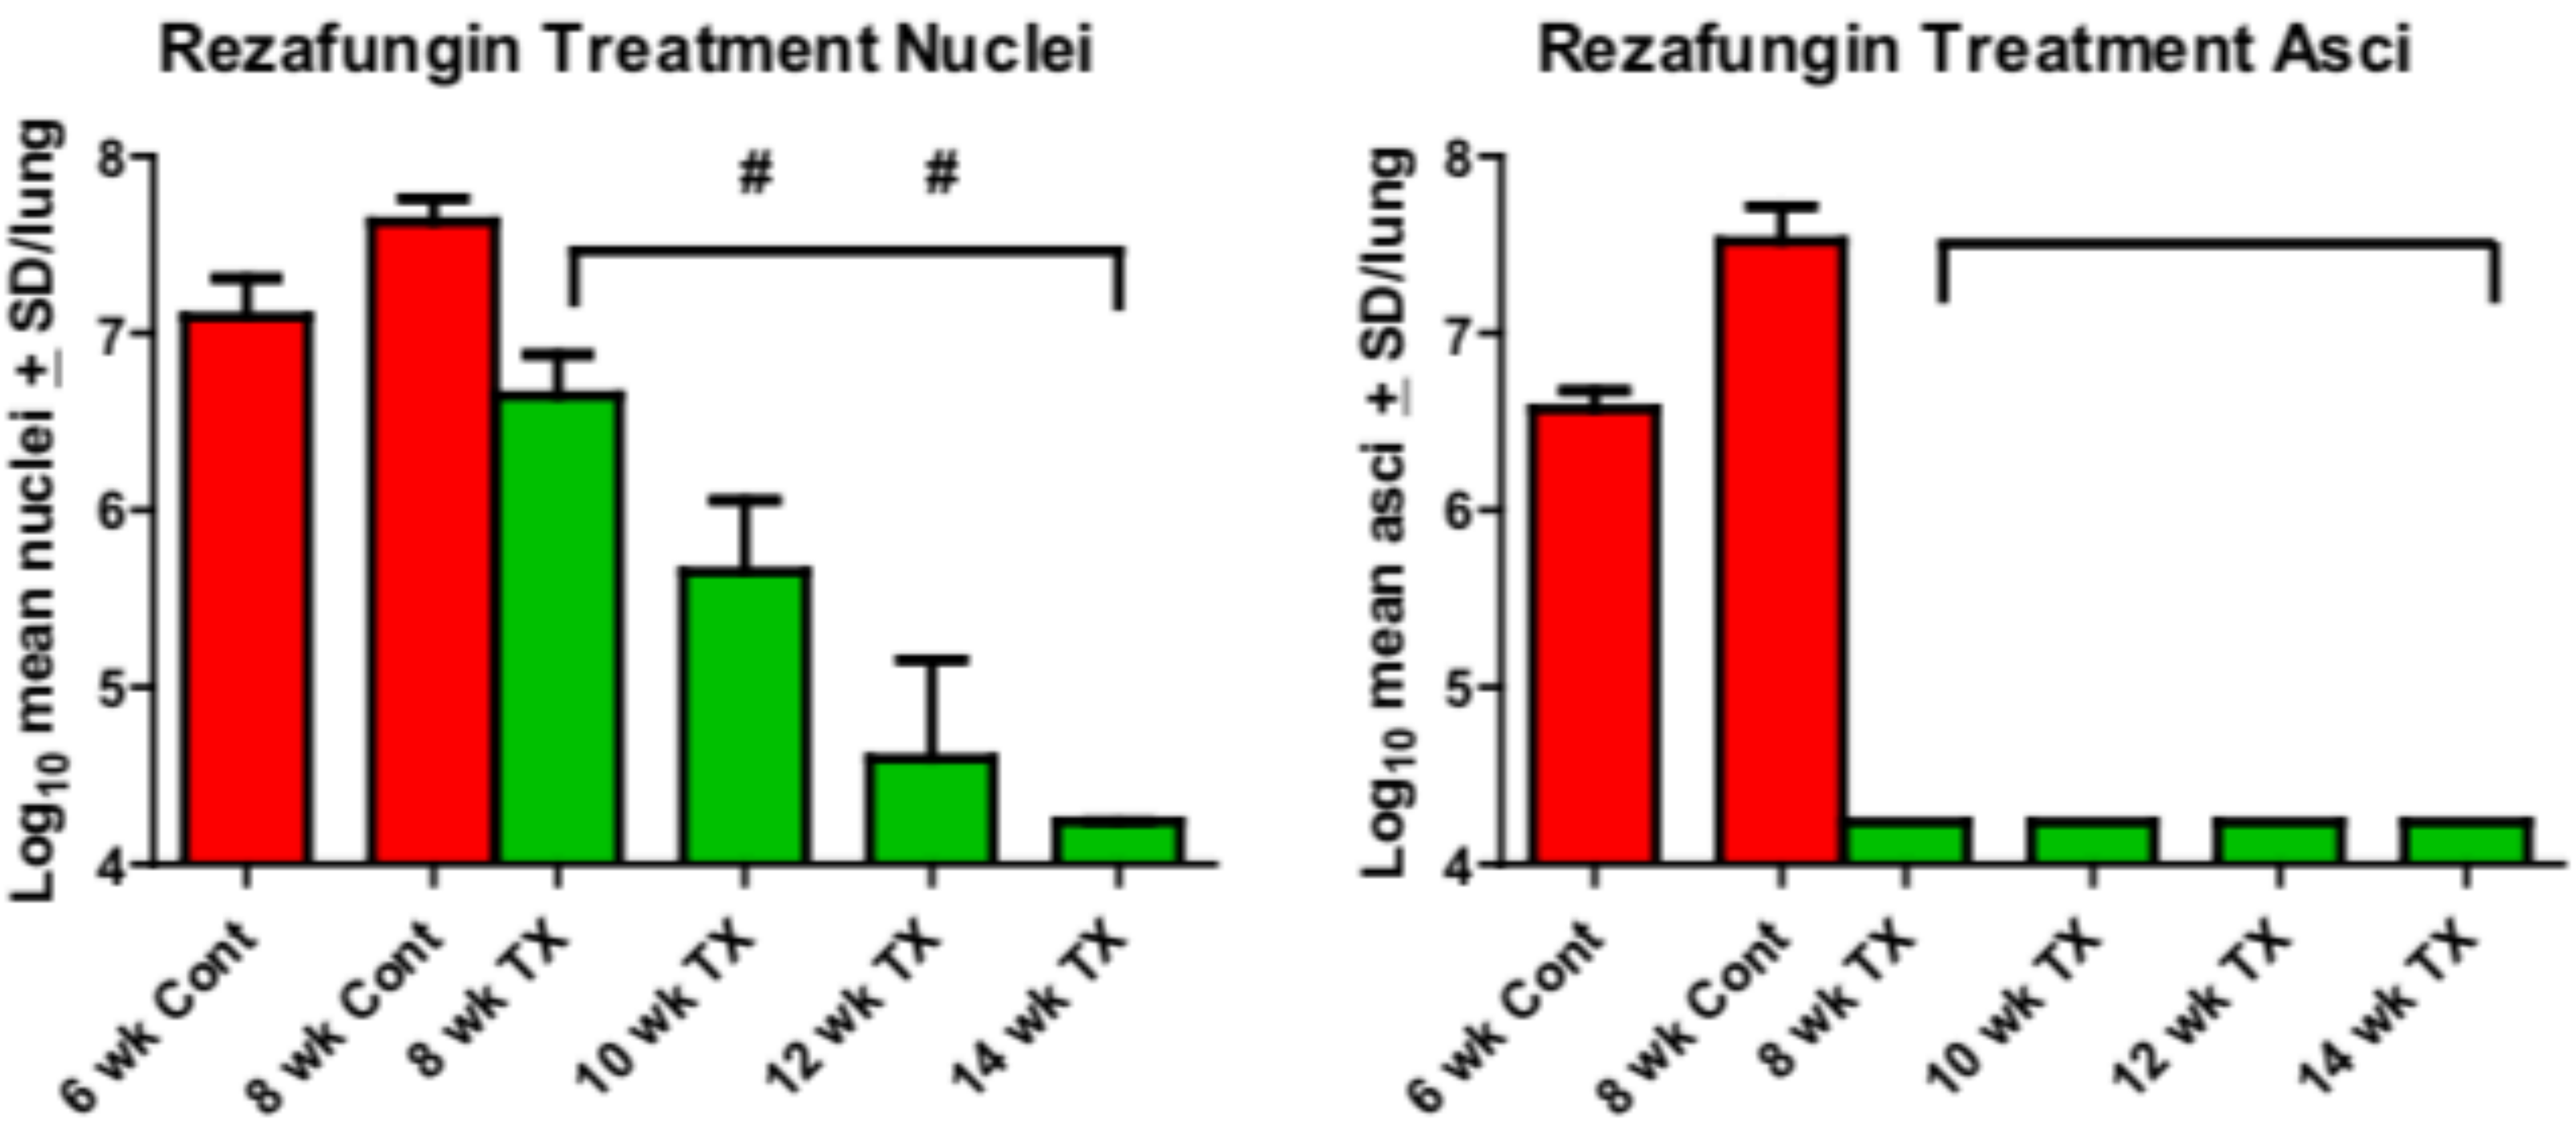
Rezafungin Treatment Nuclei
Log10 mean nuclei + SD/lung
8
7
6
5
4
#
6 wk Cont
8 wk Cont
8 wk TX
10 wk TX
12 wk TX
14 wk TX
Rezafungin Treatment Asci
Log10 mean asci + SD/lung
8
7
6
5
4
6 wk Cont
8 wk Cont
8 wk TX
10 wk TX
12 wk TX
14 wk TX

Cont=vehicle-treated negative controls (red bars). TX=rezafungin-treated with 20 mg/kg/day (green bars).
Y-axis denotes limit of detection=4.2; bracket denotes significant difference from 8-week controls; # denotes significant difference from previous week timepoint (p≤0.05).

CONCLUSIONS

- Rezafungin administered for up to 8 weeks resulted in eradication of both asci and nuclei in immunosuppressed mice infected with *P. murina*
- Eradication of nuclei by rezafungin in the absence of asci indicates that *P. murina* was unable to persist via asexual reproduction
- These results demonstrate evidence of rezafungin efficacy against *Pneumocystis* in vivo and support its ongoing clinical development for prevention of IFD

REFERENCES

1. Cordonnier et al. *J Antimicrob Chemother.* 2016;71:2379-85.
2. Redjoul et al. *Bone Marrow Transplant*. 2019;54:1082-1088.
3. Cushion et al. *PLoS One*. 2010;5:e8524.
4. Cushion et al. EBMT 2018.
5. Miesel et al. *AAC*. 2020; doi:10.1128/AAC.01992-20.

DISCLOSURES / ACKNOWLEDGEMENTS

T.S. is an employee and shareholder of Cidara Therapeutics. This study was funded by Cidara Therapeutics. Editorial support was provided by T. Chung (Scribant Medical) and funded by Cidara Therapeutics.